QUILT 3.076 Phase 1 Study of Memory-Like Cytokine-Enriched Natural Killer (M-CENK) Cells Plus NAI (N-803) In Locally Advanced or Metastatic Solid Tumors (The Cancer BioShield™)

Chaitali Nangia[1], Tara Seery[1], Katayoun Moini[1], Rebecca E. Blanton[2], Kathleen Adlard[2], Azita Nourani[2], Patricia Spilman[2], Paul Bhar[2], Hui Zhang[2], Lennie Sender[2], Sandeep Reddy[2], Patrick Soon-Shiong[2]

[1]Chan Soon-Shiong Institute for Medicine, El Segundo, CA; [2]ImmunityBio, Inc., Culver City, CA.

2025 ASCO ANNUAL MEETING

Background

Lymphopenia, characterized as low Absolute Lymphocyte Count (ALC), defined as below 1,000 lymphocyte cells/µL in blood. Currently, no treatment for lymphopenia exists. Low levels of **lymphocyte subsets of NK cells and T cells** contribute to poor prognosis and early mortality in patients with cancer. [2] *Lymphopenia and treatment induced lymphopenia (from chemotherapy, radiation, and immunotherapy) has been poorly recognized since no treatment to reverse lymphopenia existed to date until the development of NAI, an IL-15 superagonist (in-vivo) and infusion of M-ceNK (ex-vivo).* [5]

For the first time, the **in-vivo** protection of lymphocytes (via subcutaneous injection of NAI) and the **ex-vivo** protection of the collapse of the immune system (via production of M-CENK), presents a paradigm change in the treatment of cancer. We postulate that the fundamental root cause of early death in patients with cancer across all tumor types is the collapse of the immune system and that cancer is a symptom. **Lymphopenia is the disease and cancer is the symptom.** Survival is closely associated with lymphopenia and ALC, with patients who develop lymphopenia due to the cancer itself, or as a consequence of standard-of-care radiation (SBRT) or chemotherapy, having shorter survival than those with ALC in the normal range [1].

Natural Killer (NK) cells, key elements of lymphocytes subsets, **a**re the immune system's first line of defense against infection and transformed cells that may progress to cancer if they evade immune surveillance [2].

Memory-Like Cytokine-Enriched NK (M-CENK) cells generated from the patient's own PBMC-derived NK cells stimulated *ex-vivo* with IL-12, IL-18, and IL-15 superagonist nogapendekin alfa inbakicept (NAI), overcomes the collapse of the immune system across all tumor types [3].

M-CENK cells express elevated IFN-γ and granzyme B compared to healthy donor NK cells, and display toxicity against multiple tumor cell lines [4].

Cohort 1 (N=40) → Subjects Newly diagnosed with solid tumors who have not received treatment
↓
Apheresis Collection & Cryopreservation of Mononuclear Cell Aliquots
↓
Subjects will receive standard of care therapy + M-CENK

Cohort 2 (N=21) → Subjects with relapsed/refractory solid tumors who have progressive disease after receiving ≥2 prior therapies
↓
Apheresis Collection & Cryopreservation of Mononuclear Cell Aliquots
↓
Subjects receive ~4 weeks of disease-specific therapy + M-CENK

Methods

The Cancer BioShield™ (In-Vivo & Ex-Vivo)

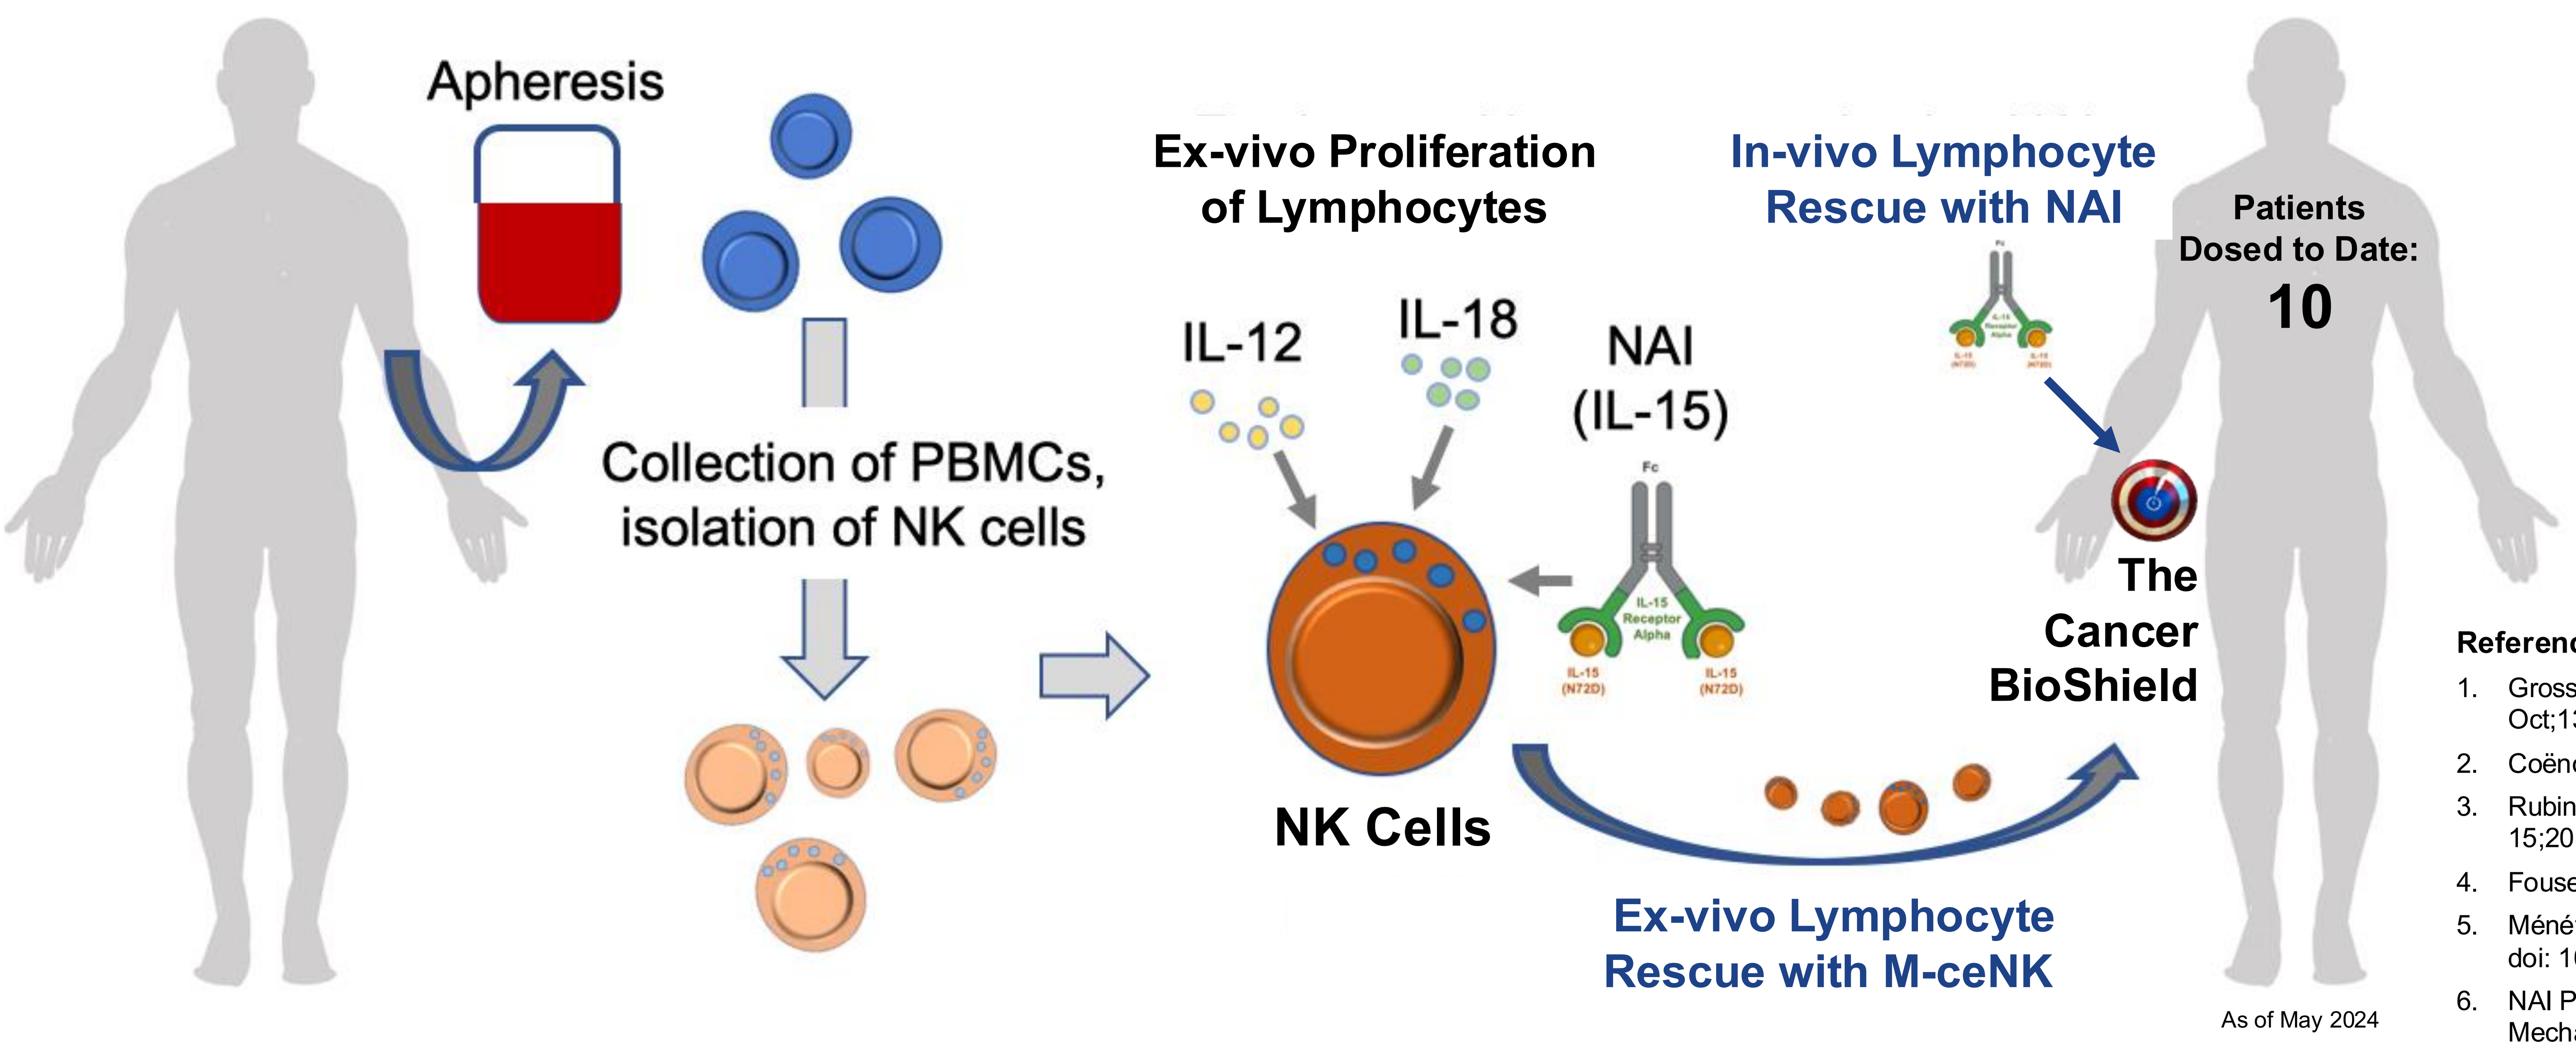
Apheresis
Collection of PBMCs, isolation of NK cells
Ex-vivo Proliferation of Lymphocytes
IL-12
IL-18
NAI (IL-15)
NK Cells
In-vivo Lymphocyte Rescue with NAI
Patients Dosed to Date:
10
The Cancer BioShield
Ex-vivo Lymphocyte Rescue with M-ceNK
As of May 2024

Download PDF & Related Articles

ASCO 2025

Info@ImmunityBio.com

References

1. Grossman, et al. *J Natl Compr Canc Netw*. 2015 Oct;13(10):1225.
2. Coënon, et al.. *Cell Death & Disease* 2024,15: 614.
3. Rubinstein, et al. Clinical Trial J Immunol. 2022 Mar 15;208(6):1362-1370.
4. Fousek, et al. J Thorac Oncol. 2023 Mar;18(3):350.
5. Ménétrier-Caux C,et al. J Immunother Cancer;;7(1):85. doi: 10.1186/s40425-019-0549-5.
6. NAI Package Insert, FDA April 2024 – Section 12.1 Mechanism of Action

Phase 1 First-in-Human Study QUILT-3.076 (NCT04898543) assesses the safety and preliminary efficacy of M-CENK cells plus NAI in participants with locally advanced or metastatic solid tumors.

- **Cohort 1** (up to N=40) enrolls participants with newly diagnosed, 1st line solid tumors.
- **Cohort 2** (up to N=21) enrolls participants with relapsed or refractory solid tumors who progressed after ≥ 2 lines of therapy.

M-CENK cells are administered by infusion (N=10 Enrolled to Date) weekly up to 10 times and NAI SC for up to 5 doses every 2 weeks prior to every other dose of M-CENK cells.

Primary Objective: Safety
TEAEs, SAEs, & clinically significant changes in laboratory tests & vital signs. Toxicities graded using CTCAE v5.0 or a specified grading system for CRS

Efficacy Objectives in Cohort 2B is ORR (RECIST v1.1 & iRECIST criteria) PFS, OS using KM methods.

Secondary Objectives are Evaluation of M-CENK Infusion + NAI Subcutaneous:

SUMMARY TO DATE:

- Patient infusions to date: N=10 (as of May 2025). All infusion performed in an outpatient setting
- Cancer types (2nd line & greater): Breast (N=4), Colon (N=1), Duodenum (N=1), Renal (N=1), Pancreatic (N=1), Rectal (N=1), Osteosarcoma (N=1)
- Range of M-ceNK infusions (2 to 5 bags infused) with NAI subcutaneously
- Safety: Zero (0%) TRAE Grade 4 or 5. Zero (0%) cytokine storm