Generation of Cytokine-Induced Memory-Like PD-L1 t-haNK™ (NK-92®) Cells

*Dustin Elwood, Devapriya Segaran, Joseph Thibault, Vidya Godbole, Manju Saxena, Patrick Soon-Shiong, Hans Klingemann, Laurent Boissel
ImmunityBio, Inc. - 9920 Jefferson Blvd, Culver City, CA – USA *dustin.elwood@immunitybio.com

Abstract

Cytokine-induced memory-like NK cells have garnered a lot of attention as cellular therapeutic agents due to their much-increased lifespan in patients and their enhanced memory response upon re-challenge by cytokines or target cells.

We show here that the Natural Killer cell line NK-92®[1] or 'activated' NK (aNK™) can acquire activation as well as a memory-like (ML) phenotype upon overnight induction with a cytokine cocktail including IL-12, IL-18, and the IL-15 superagonist N-803, as evidenced by increased steady-state IFN-γ secretion (>50-fold increase) and up-regulation of CD25. Further, NK-92 cells engineered to express a high-affinity Fc receptor and an anti-PD-L1 Chimeric Antigen Receptor (CAR) (PD-L1 t-haNK™) show similar characteristics. Exposure of the NK-resistant cells lines THP-1 and SK-ES-1 to culture supernatant from memory-like PD-L1 t-haNK triggered expression of PD-L1 on their surface, rendering them highly sensitive to PD-L1 CAR-mediated targeted killing by PD-L1 t-haNK cells. Target cell upregulation of ICAM-1 was also observed, potentially opening tumor targets up to general immune engagement.

Infusion of a cellular therapy product that constitutively secretes high quantities of IFN-γ may cause detrimental side effects. Memory-like PD-L1 t-haNK cells can circumvent this problem through their memory recall ability and deliver increased amounts of IFN-γ specifically at the tumor site when locally engaging with tumor cells. This increase would then drive upregulation of PD-L1 expression on neighboring tumor cells, creating an expanding wave of targets susceptible to killing by PD-L1 t-haNK cells. This approach provides an avenue to defeat immunologically cold cancers.

Abstract
#894

Introduction

- Memory-like NK cells present a valuable therapeutic option due to their safety and their effectiveness through enhanced memory recall response[2,3].
- A key part of this memory recall feature is the increased secretion of interferon gamma (IFN-γ). This potent immune stimulator can trigger upregulation of MHC-I molecule expression on the surface of tumor cells, thereby pushing the tumor towards an immunologically "hot" status as well as increase surface ICAM-1 expression enabling engagement from NK cells.
- IFN-γ also triggers expression of the immune checkpoint molecule PD-L1[4]. While PD-L1 expression can act to disable some immune responses, a PD-L1 CAR T-cell or NK cell can take advantage of such defenses[5].
- Generation of a memory-like NK-92-based cell line could allow a local recall of these enhanced activation features upon contact with tumor cells while avoiding the dangers of systemic IFN-γ production.

Materials and Methods

Memory-like (ML) cytokine stimulation cocktail: IL-12, IL-18, and N-803 were added to cell culture medium (X-VIVO™ 10 + 5% human serum) for 14-16 hours followed by return to normal cell culture medium (500 IU/mL rhIL-2). Time following ML stimulation are described in figure illustrations.

Intracellular flow cytometry: Fixation by BD Cytofix/Cytoperm™ kit. IFN-γ antibody Biolegend or Miltenyi Biotec.

Extracellular flow cytometry: Antibodies: MHC class I "HLA A, B, C PE" Biolegend, PD-L1 APC BD Biosciences, CD54 (ICAM1) BD. Live/dead using DAPI Miltenyi. All staining prepared in Biolegend Cell Staining Buffer.

IFNγ ELISA: R&D Systems™ Human IFN-gamma Quantikine ELISA Kit, Fisher Scientific. Cell Culture supernatants run at 2- and 80-fold dilutions. Time course followed Figure 1c illustration.

Cytotoxicity Assays: 4 hours effector-to-target (E:T) coincubation at 37°C using PKH67GL stain (Sigma PKH67GL-1 KT) to identify target cells and propidium iodide staining for viability.

CyTOF: Helios CyTOF N10410 (Standard Bio). MaxPar Direct Immune Profiling Assay Foil Packet, Standard Bio.

References

1. Klingemann, H. (2023) The NK-92 cell line-30 years later: its impact on natural killer cell research and treatment of cancer. *Cytotherapy*, 25(5):451-457. doi.org/10.1016/j.jcyt.2022.12.003
2. Romee, R., *et. al*. (2012) Cytokine activation induces human memory-like NK cells. *Blood,* 120(24):4751-60. doi: 10.1182/blood-2012-04-419283
3. Marin, N. D., *et. al.* (2021) Memory-like Differentiation Enhances NK Cell Responses to Melanoma. *Clin Cancer Res,* 27:4859–69 doi: 10.1158/1078-0432.CCR-21-0851.
4. Garcia-Diaz, A., *et. al*. (2017) Interferon Receptor Signaling Pathways Regulating PD-L1 and PD-L2 Expression. *Cell Rep.* 19(6): 1189–1201. doi:10.1016/j.celrep.2017.04.031.
5. Eitler, J., *et. al.* (2024) CAR-mediated targeting of NK cells overcomes tumor immune escape caused by ICAM-1 downregulation. *J Immunother Cancer,* 12(2):e008155. doi: 10.1136/jitc-2023-008155.

Results

Memory-like (ML) cytokine stimulation results in increased memory markers and activation signals.

(A) aNK and PD-L1 t-haNK CyTOF intracellular marker expression following memory-like cytokine stimulation. Among other markers, IFN-γ is maintained by both ML stimulated NK-92-derived cells lines. NK-Panel samples taken ~16 hours post stimulation with IL-12, IL-18, and N-803. **(B)** Intracellular IFN-γ (left) and GZMB (right) show rapid increases that return to pre-stimulation levels after 48 hours. **(C)** Measurement of secreted IFN-γ by ELISA indicates that ML-aNK can display memory recall for at least 7 days post-activation upon re-challenge with K562, Raji, and THP-1 target cells (2.3, 6.9, and 3.8-fold increases after 1 week, respectively).

A.

	aNK (% Positive Cells)		PD-L1 t-haNK (% Positive Cells)	
Protein	unstim	ML stim	unstim	ML stim
CD56	99.2	99.5	97.3	98.5
CD69	81.8	68	92.4	70
HLA-DR	14.6	13.9	19.3	23.2
CD38	95.9	97.1	99.9	99.9
CD16	7.9	3.1	91.5	92.4
CD25	3.5	95.6	2.4	5.5
CD27	0.3	4	0.2	0.3
CD62L	1.8	2.1	3.5	12.1
PD-1	3.4	5.4	4.5	11.9
IFNγ	3.7	62.4	5.3	22.8
NKp30	76.7	91	77.9	92.3
NKp46	58.8	62	64.3	70.1
LAG-3	1.6	17.6	1.5	29.6
NKG2D	90.4	90.1	92.2	91.8
KIR3DL1	6.1	10.4	11.9	10.3
Ki-67	99.4	99.7	99.6	99.4
NKG2A	99.2	99.1	99.9	99.9
Granzyme B	98.5	98.3	98.8	98.7
CD57	0.8	0.6	0.4	0.5
CD94	100	99.9	100	100
Perforin	96.9	98.3	99.4	99.1
TIGIT	91	93.2	92.5	92

B.

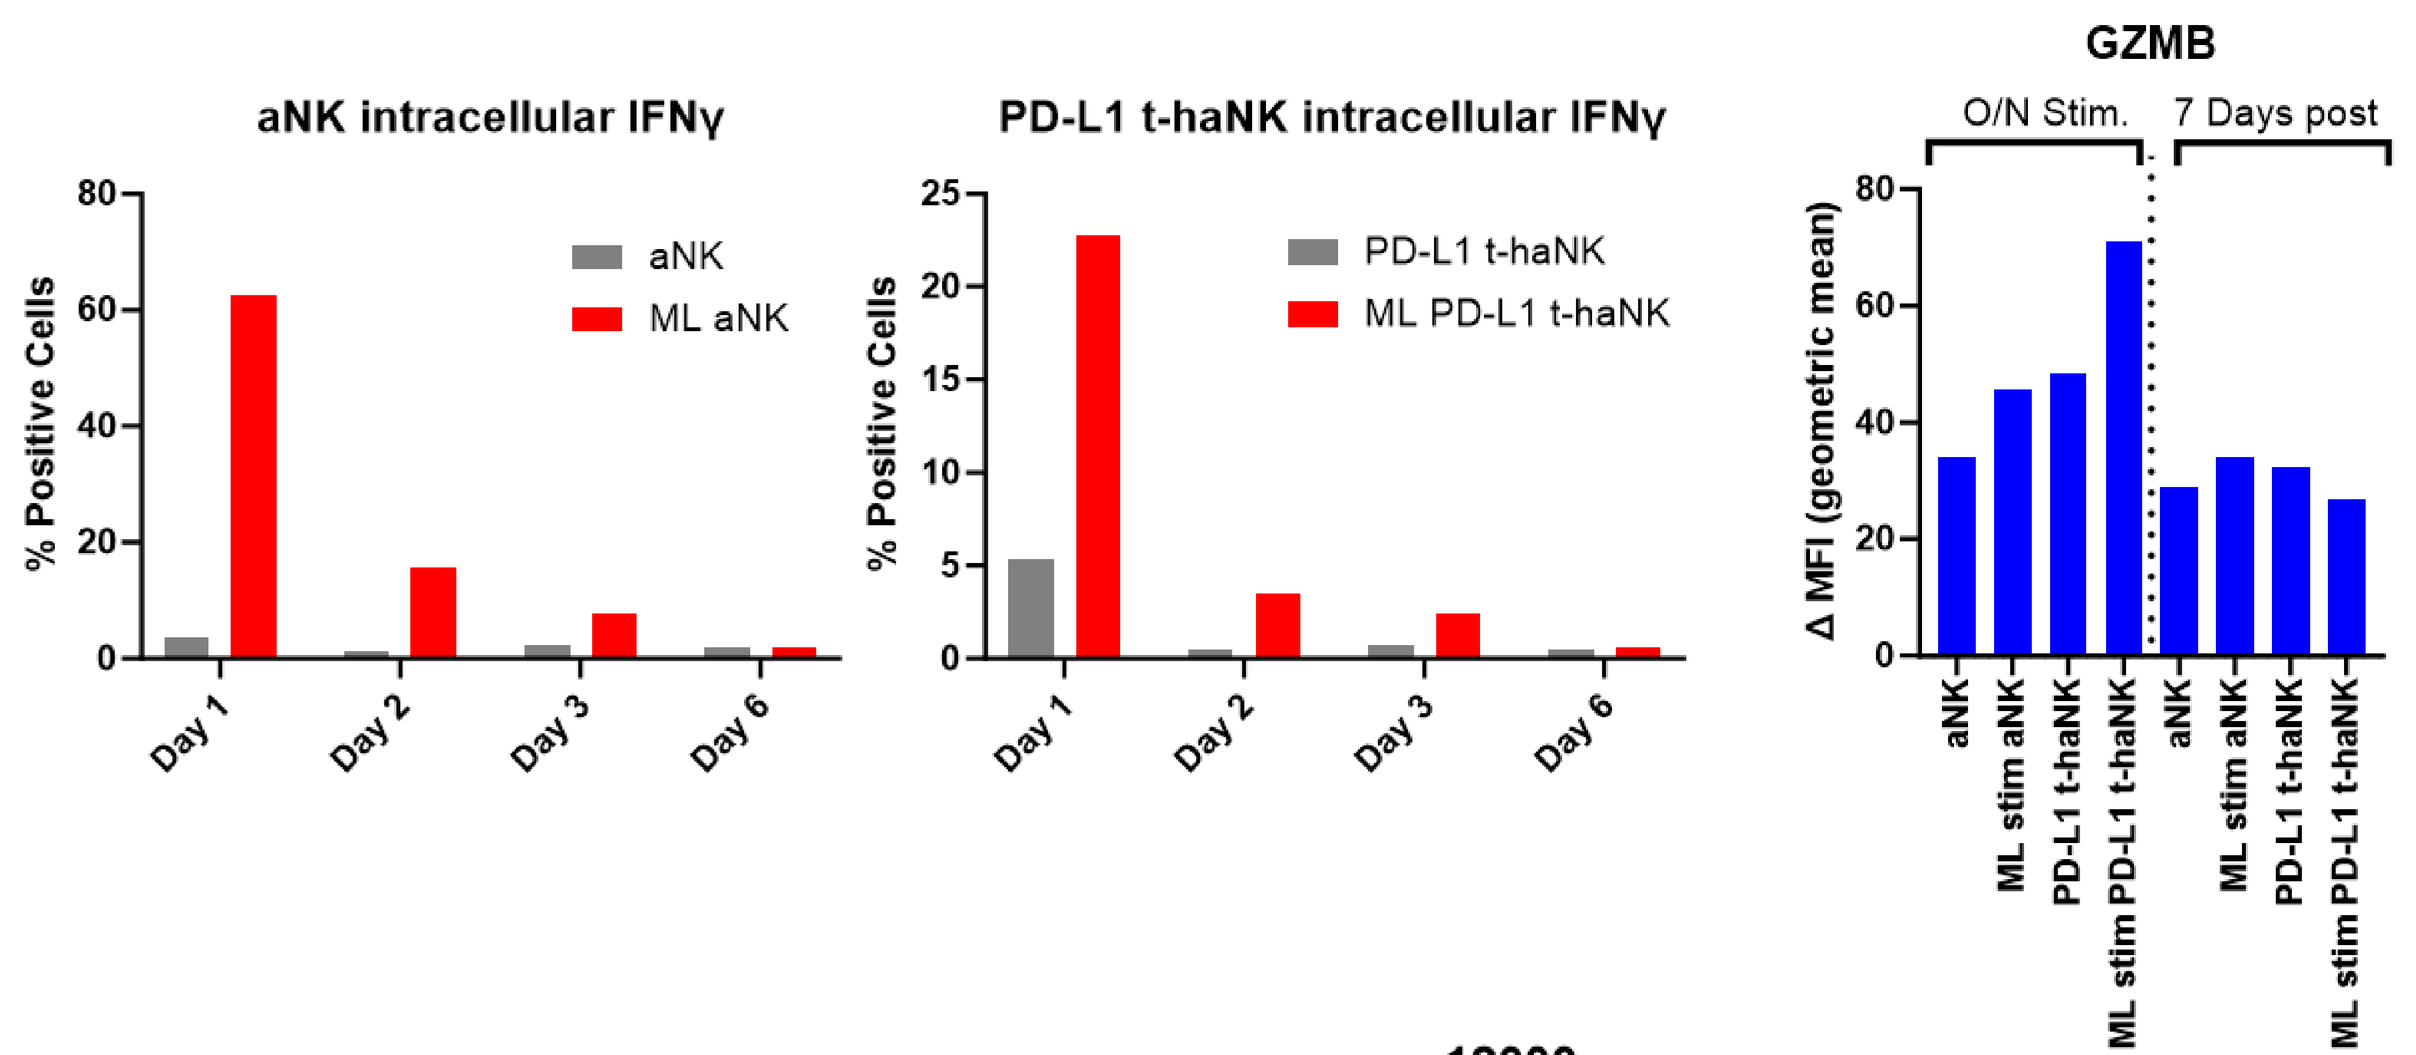
aNK intracellular IFNγ
aNK
ML aNK
% Positive Cells
Day 1
Day 2
Day 3
Day 6
PD-L1 t-haNK intracellular IFNγ
PD-L1 t-haNK
ML PD-L1 t-haNK
GZMB
O/N Stim.
7 Days post
Δ MFI (geometric mean)
aNK
ML stim aNK
PD-L1 t-haNK
ML stim PD-L1 t-haNK

C.

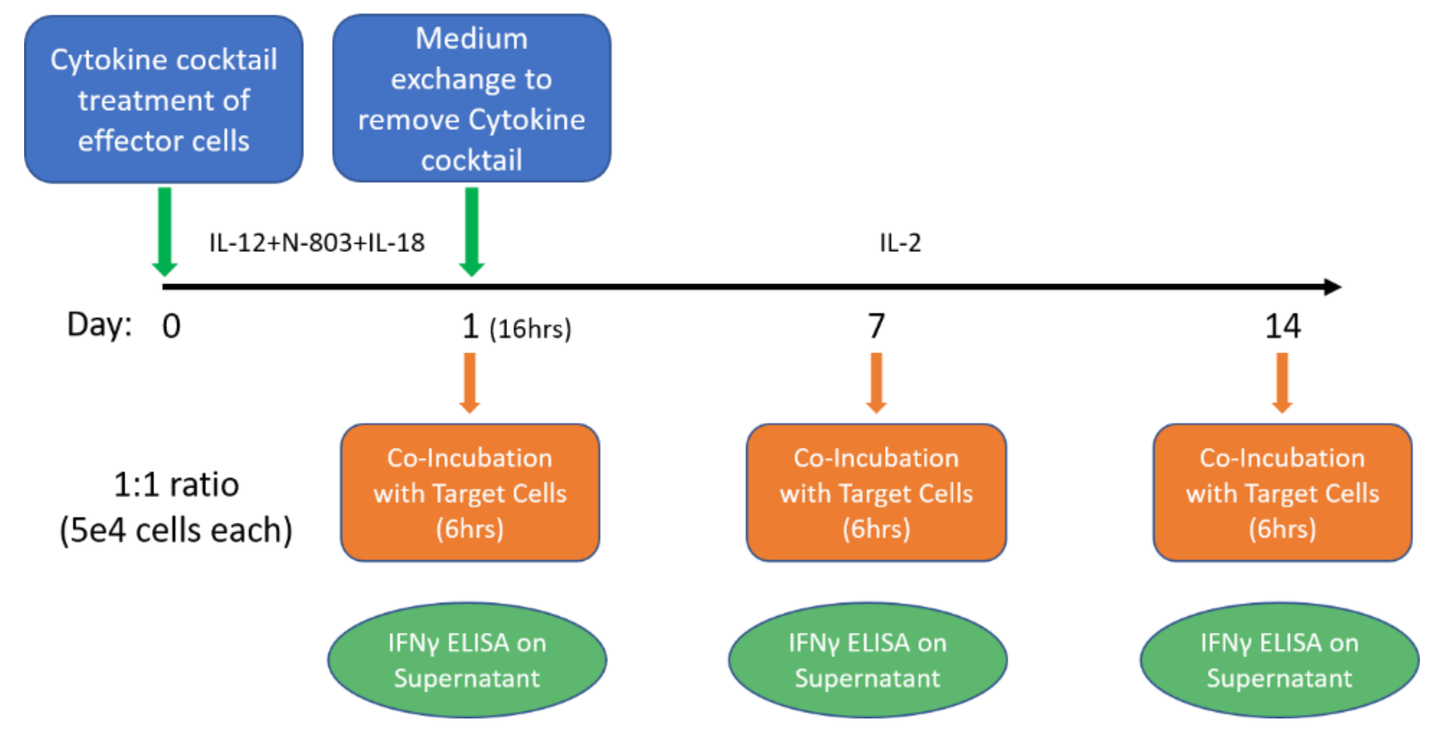
Cytokine cocktail treatment of effector cells
Medium exchange to remove Cytokine cocktail
IL-12+N-803+IL-18
IL-2
Day: 0
1 (16hrs)
7
14
1:1 ratio
(5e4 cells each)
Co-Incubation with Target Cells (6hrs)
IFNγ ELISA on Supernatant

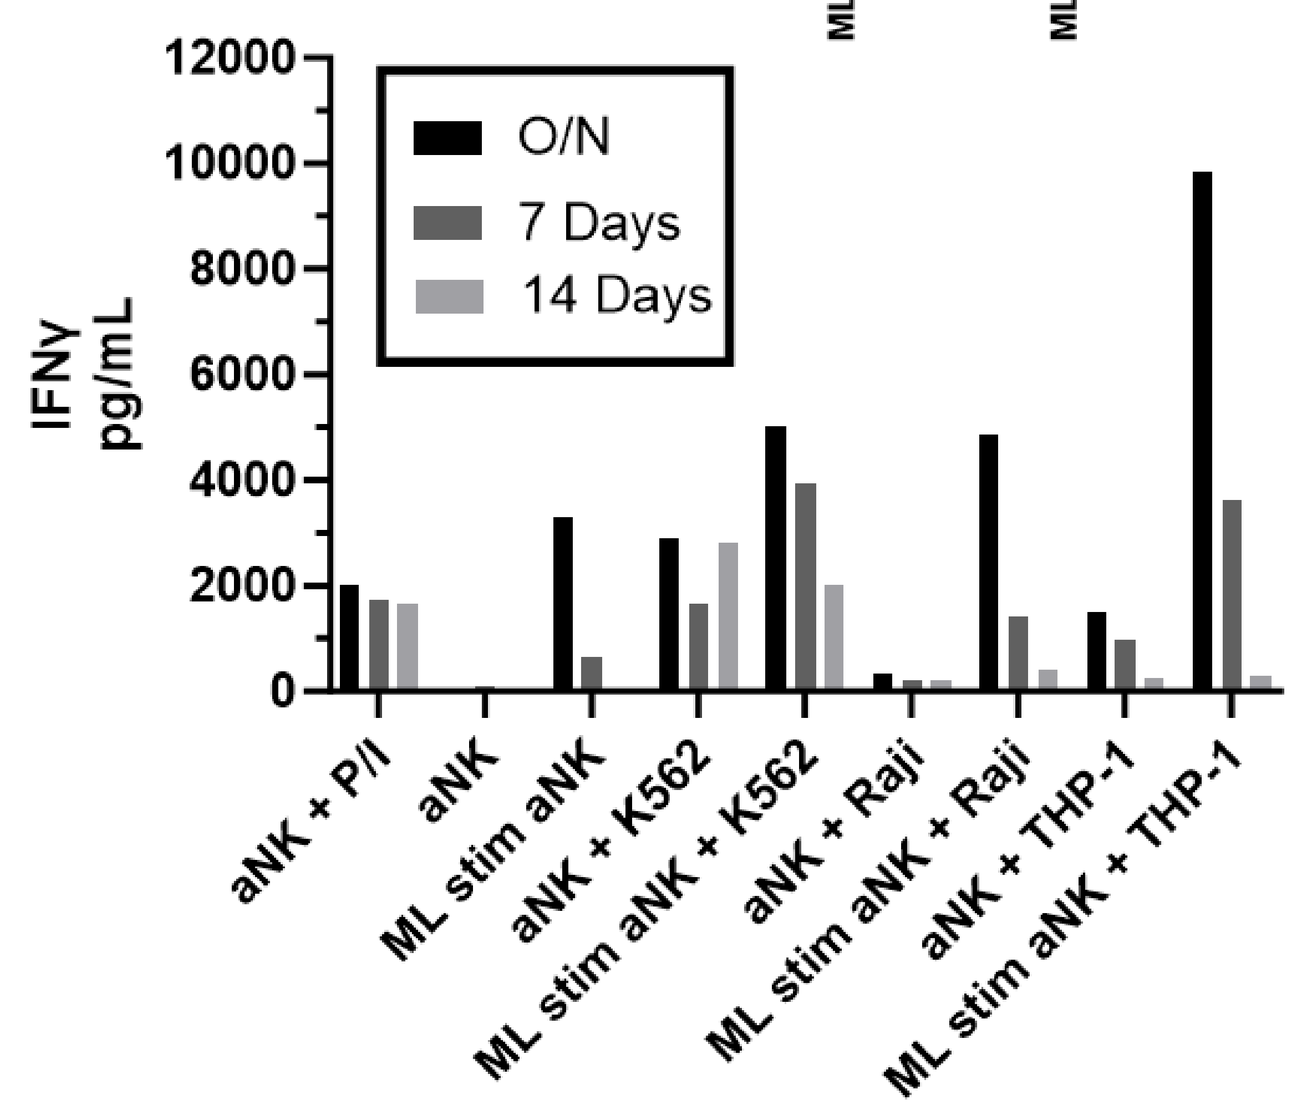
IFNγ pg/mL
O/N
7 Days
14 Days
aNK + P/I
aNK
ML stim aNK
aNK + K562
ML stim aNK + K562
aNK + Raji
ML stim aNK + Raji
aNK + THP-1
ML stim aNK + THP-1

Exposure to memory-like effector cell supernatant results in changes to target cell surface markers.

PD-L1 and ICAM-1 surface markers increase on resistant THP-1 target cells as a result of exposure to IFN-γ or ML aNK /PD-L1 t-haNK cell culture supernatant. Increased ICAM-1 can elicit increased killing by NK cells.

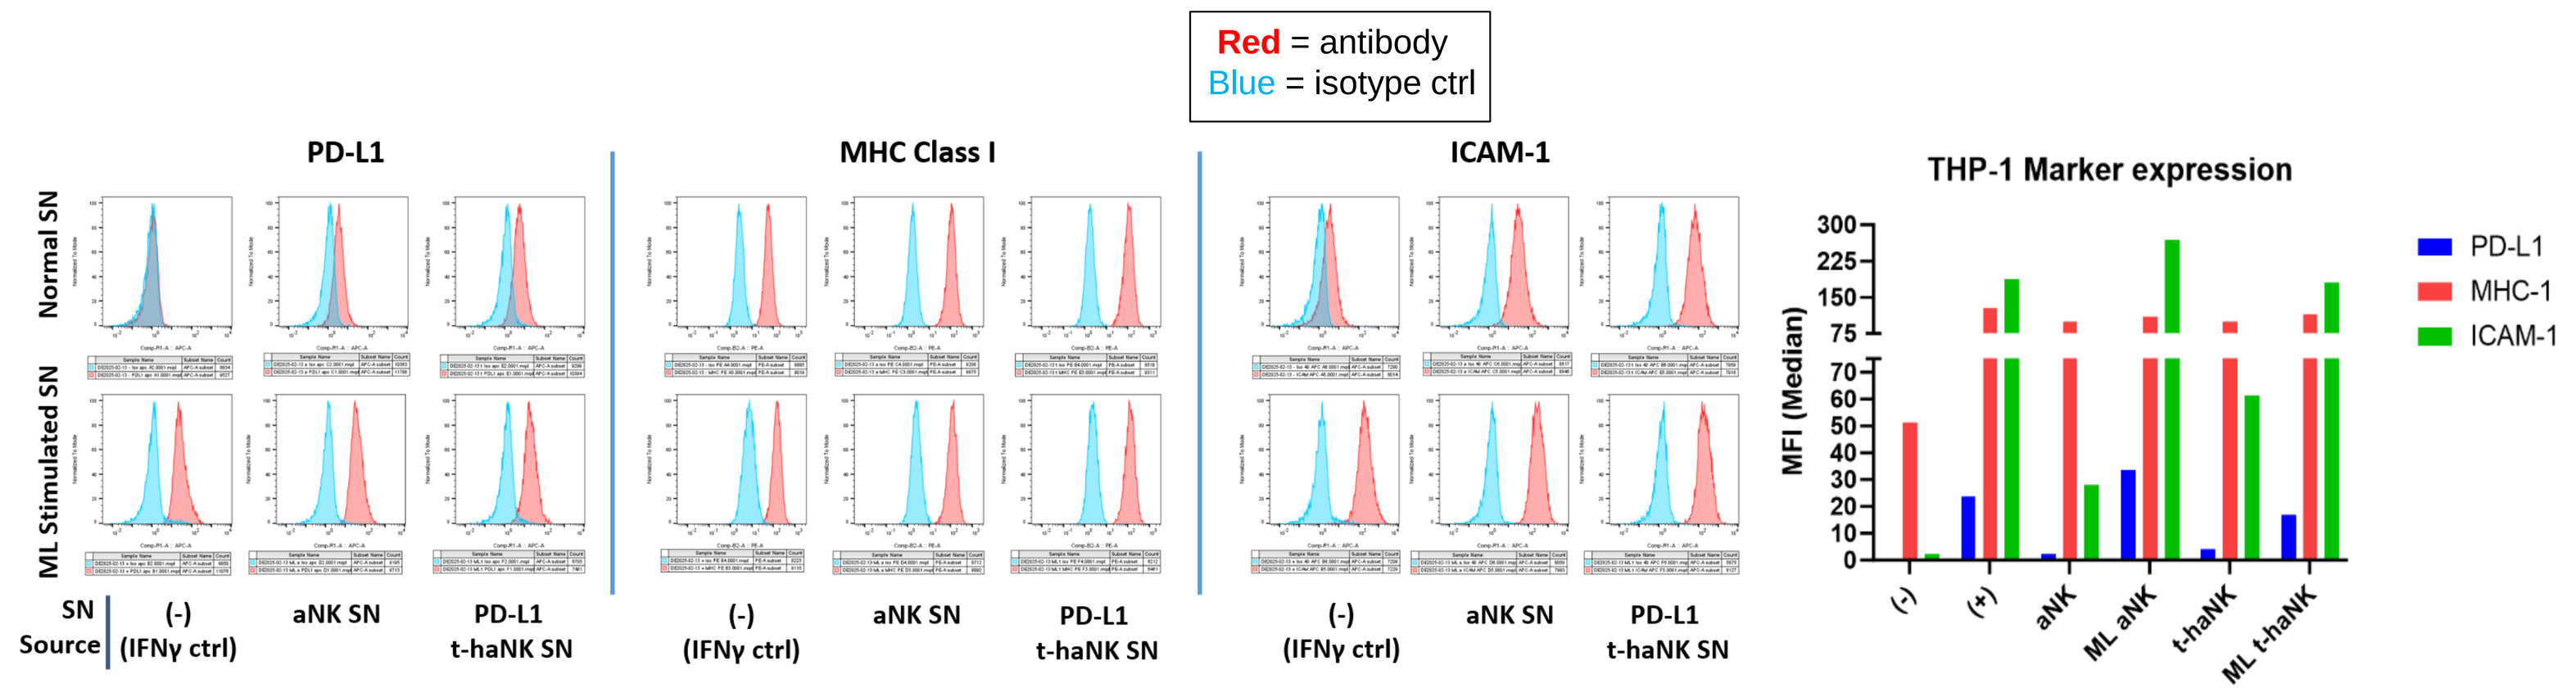
Red = antibody
Blue = isotype ctrl
PD-L1
MHC Class I
ICAM-1
Normal SN
ML Stimulated SN
SN Source
(-) (IFNγ ctrl)
aNK SN
PD-L1 t-haNK SN
THP-1 Marker expression
MFI (Median)
PD-L1
MHC-1
ICAM-1
(-)
(+)
aNK
ML aNK
t-haNK
ML t-haNK

ML PD-L1 t-haNK-secreted factors increase susceptibility of targets to killing by PD-L1 t-haNK.

THP-1 and SK-ES-1 targets treated with cell culture supernatant from ML aNK/PD-L1 t-haNK-are sensitized to PD-L1 CAR-mediated targeted killing by the PD-L1 t-haNK cells in a 4-hrs *in vitro* cytotoxicity assay. Increased PD-L1 surface expression enables direct targeting by PD-L1 t-haNK and can enable a positive feedback loop for eliminating resistant targets.

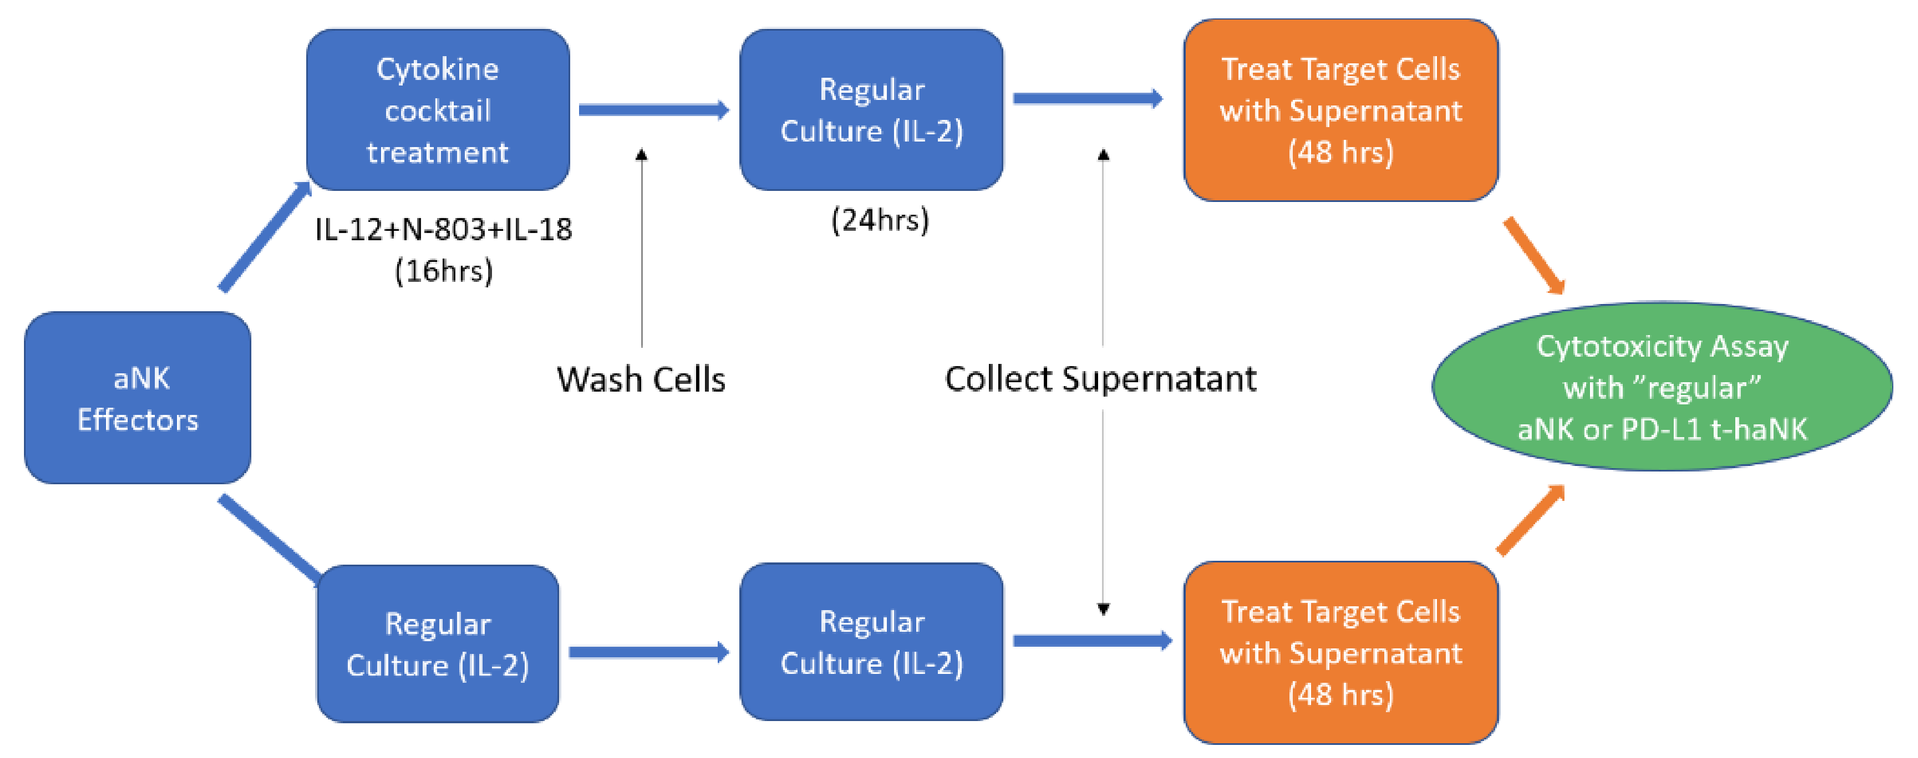
aNK Effectors
Cytokine cocktail treatment
IL-12+N-803+IL-18 (16hrs)
Regular Culture (IL-2)
(24hrs)
Treat Target Cells with Supernatant (48 hrs)
Wash Cells
Collect Supernatant
Cytotoxicity Assay with "regular" aNK or PD-L1 t-haNK

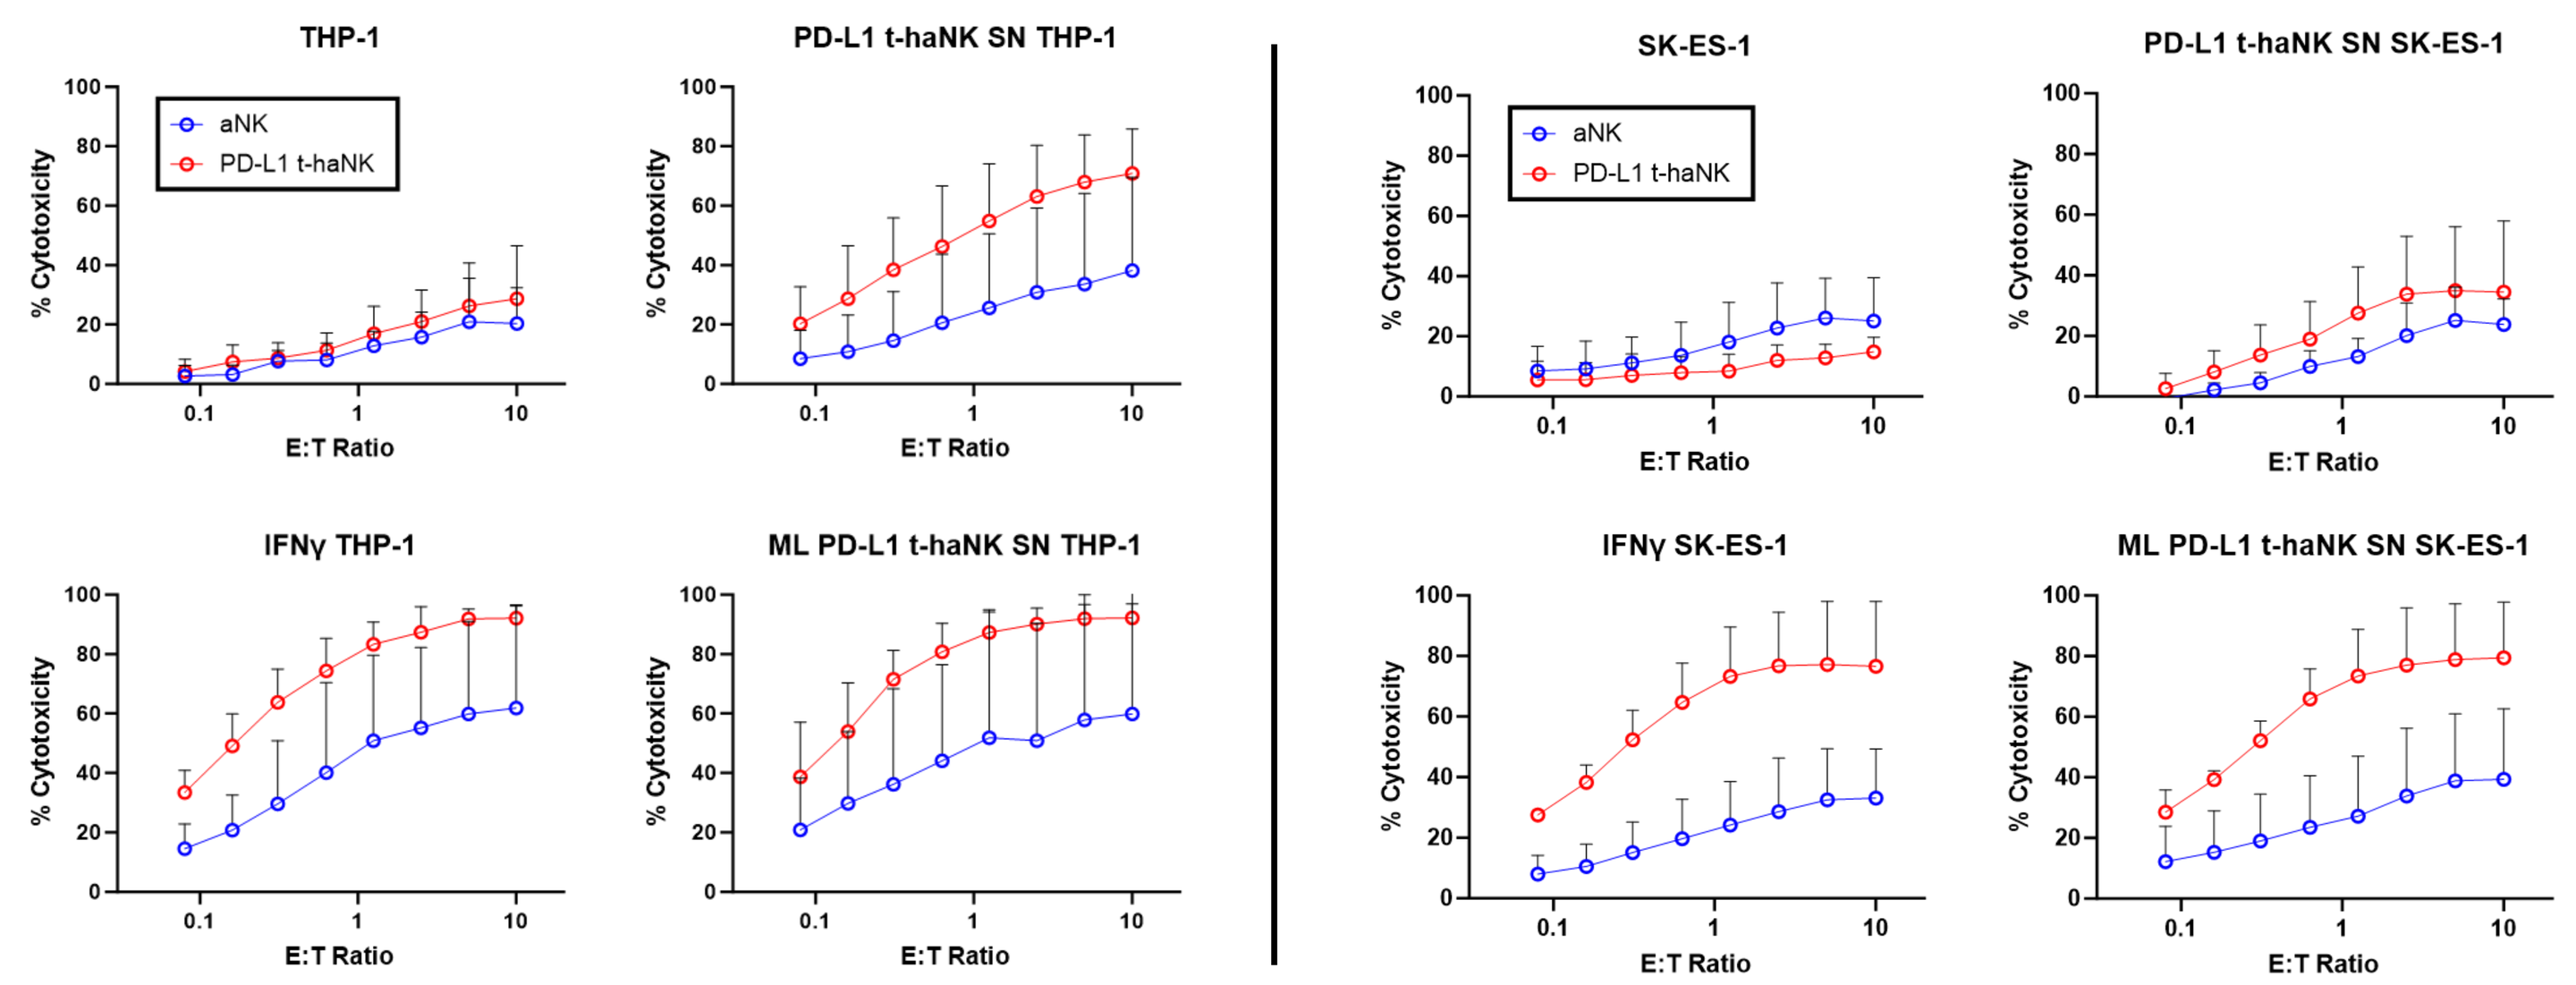
THP-1
PD-L1 t-haNK SN THP-1
IFNγ THP-1
ML PD-L1 t-haNK SN THP-1
SK-ES-1
PD-L1 t-haNK SN SK-ES-1
IFNγ SK-ES-1
ML PD-L1 t-haNK SN SK-ES-1
aNK
PD-L1 t-haNK
% Cytotoxicity
E:T Ratio

Discussion and Conclusions

The work presented here demonstrates a way to generate an off-the-shelf NK cell therapy that could deliver IFN-γ to tumor sites and avoid the dangers of systemic IFN-γ exposure to the patient. By using memory-like (ML) PD-L1 t-haNK cells, we are able to engage resistant tumor types that express the immune checkpoint PD-L1. This positive feedback loop of target engagement and elimination coupled with induction of PD-L1 on nearby tumor cells builds a tumor microenvironment (TME) that favors therapeutic success. Further induction of ICAM-1 allows improved engagement with LFA-1 on PD-L1 t-haNK as well as other immune cells. ML PD-L1 t-haNK with extended half-life in patients is being developed and would constitute a promising cellular therapy against resistant cancers.